Development Of An Upstream Process And Analytics For AAV Manufacturing

Melissa Gosse[1], Shibi Likhite[2], Desyree Jesus[1], Kathrin Meyer[2], and Susan D'Costa[1]

[1] Alcyone Therapeutics, Lowell, MA, 01852; [2] Center for Gene Therapy Abigail Wexner Research Institute, Nationwide Children's Hospital, Columbus, OH, 43205

Background

Development of a robust and scalable manufacturing process early in a product's lifecycle is critical since gene therapy trials are often accelerated. However, establishing such a process often requires a significant amount of time and investment. Although several upstream processes are available for AAV production, optimizing the one with most productivity, scalability and best cost-of-goods for commercial production remains a challenge. With the choice between the basic processes of suspension and adherent, a variety of other factors like scalability, yield, use of animal-derived components, quality target product profile and even licensing requirements need to be considered to minimize the target timing to get to the clinic.

For a successful process, it is also crucial to begin analytical development of product specific assays either before or in parallel to process development, the most critical of assays being the dosing (vg) assay and the potency assay. Early analytical development is imperative to not only help progress process development, but also to ensure assays will be ready for qualification and timely release of product.

The transgene construct for our current process utilizes self complementary AAV serotype 9 with the actual transgene containing repetitive elements and secondary structure. These qualities make it challenging to develop a robust and consistent vg titer assay. This poster will highlight the parameters optimized to develop both a dosing and expression assay. In addition, this poster will focus on parameters evaluated to build out a robust platform upstream process that is scalable and easily transferrable between multiple systems.

Analytical Development

Vg titer assay

In order to develop a robust ddPCR-based vg titer assay for a challenging GOI that contains repetitive sequences and secondary structures, different parameters in the flow diagram below (red circles) were optimized. The parameter was considered optimized if the percent of rain was less than 2% (black dashed line in graphs below).

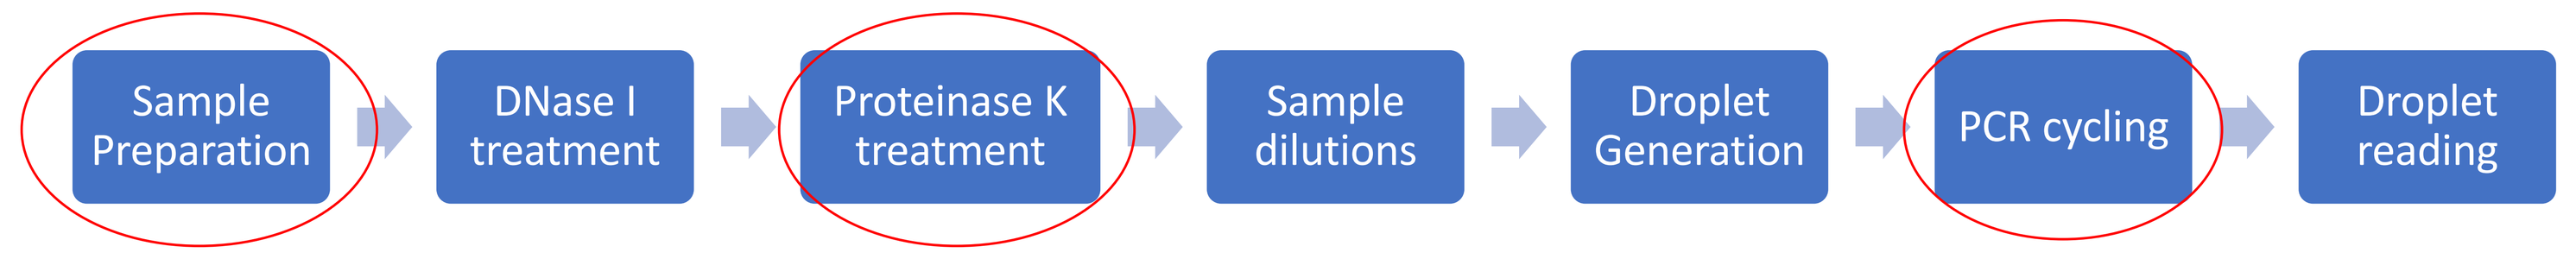

Sample Preparation
DNase I treatment
Proteinase K treatment
Sample dilutions
Droplet Generation
PCR cycling
Droplet reading

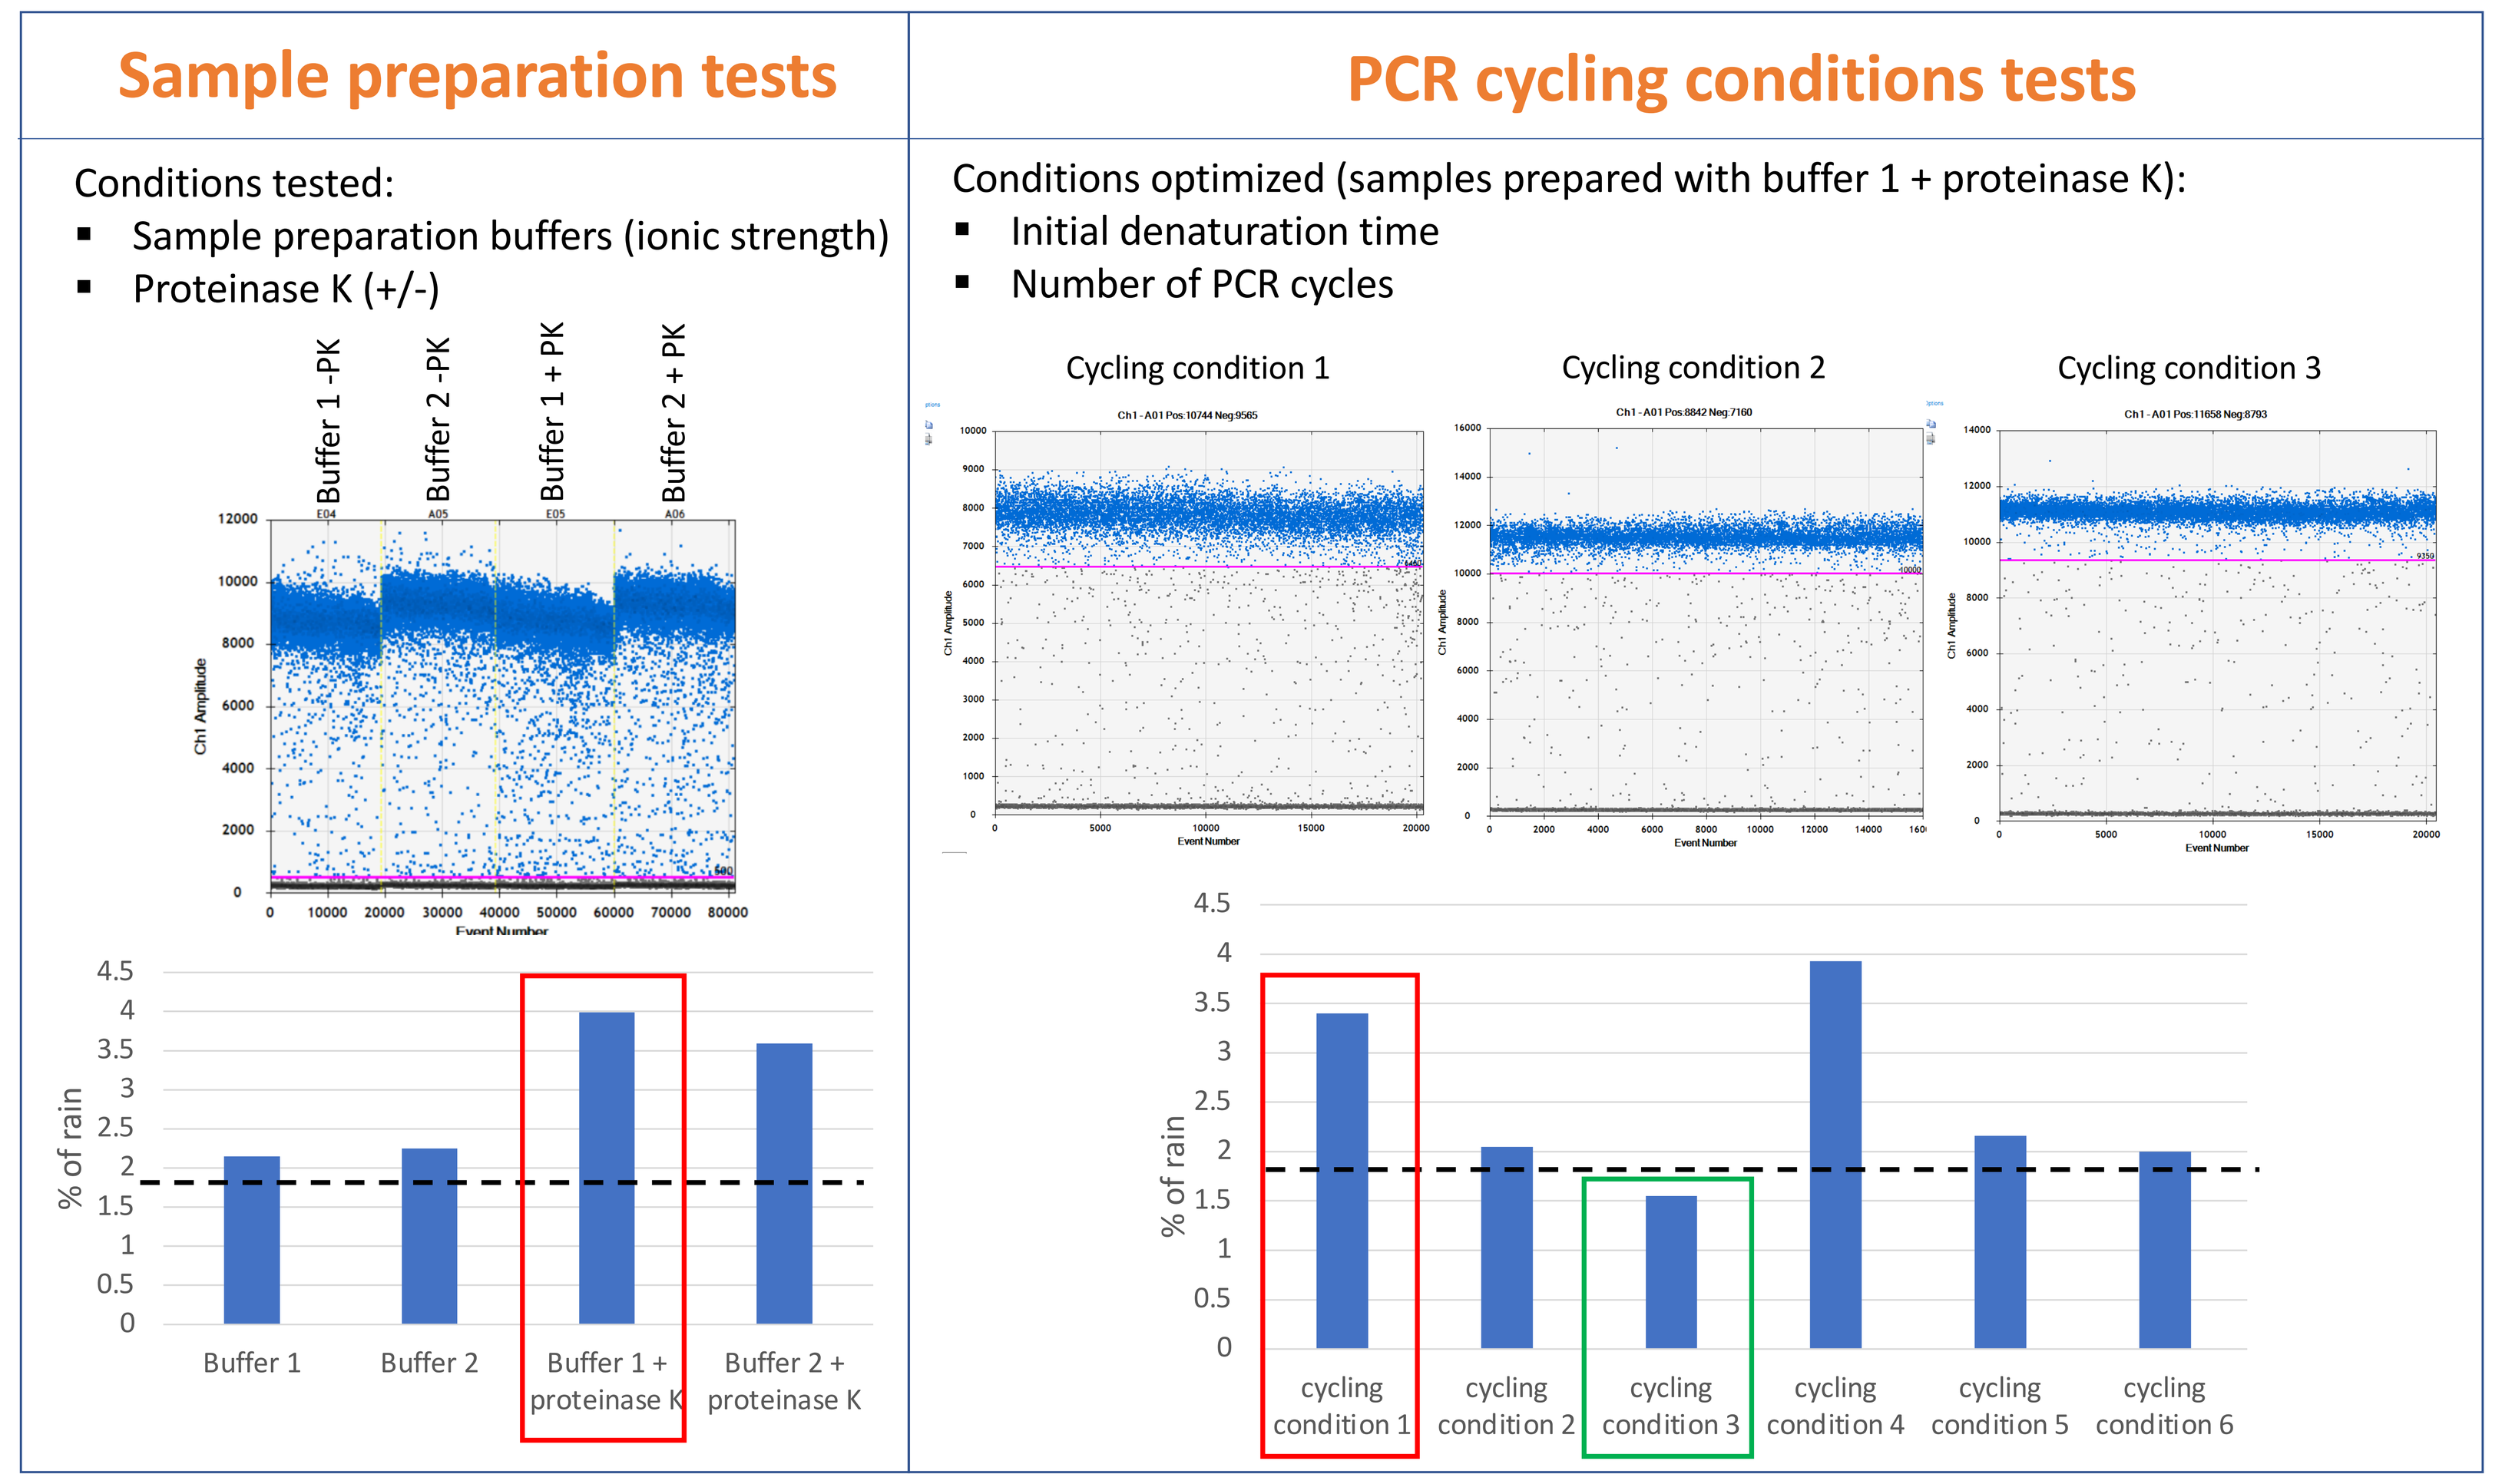

Sample preparation tests
Conditions tested:
Sample preparation buffers (ionic strength)
Proteinase K (+/-)
Buffer 1 -PK
Buffer 2 -PK
Buffer 1 + PK
Buffer 2 + PK
Ch1 Amplitude
Event Number
% of rain
Buffer 1
Buffer 2
Buffer 1 + proteinase K
Buffer 2 + proteinase K
PCR cycling conditions tests
Conditions optimized (samples prepared with buffer 1 + proteinase K):
Initial denaturation time
Number of PCR cycles
Cycling condition 1
Cycling condition 2
Cycling condition 3
% of rain
cycling condition 1
cycling condition 2
cycling condition 3
cycling condition 4
cycling condition 5
cycling condition 6

FIGURE 1: Representative ddPCR graphs of rain observed in few selected conditions are shown above. These data were transformed into bar graphs to quantitate the percent of rain observed for the different conditions tested. The original protocol is highlighted in red rectangles. The final selected protocol is highlighted in a green rectangle (no changes were made to the sample preparation conditions in the final protocol).

ALCYONE THERAPEUTICS

Potency assay – Expression of transgene (short RNA)

The transgene is a short RNA that will not be translated into protein but utilized to "sponge" up a miRNA of interest. This product is intended for use as a "pipeline in a product" and will be leveraged for multiple indications that are impacted by this mechanism of action. Alcyone's platform strategy is to use an expression assay for early phase and a functional assay, showing the mechanism of action of the vector, at later phases. The expression assay as described in the flow diagram below determines RNA amounts by RT-ddPCR normalized to a housekeeping gene.

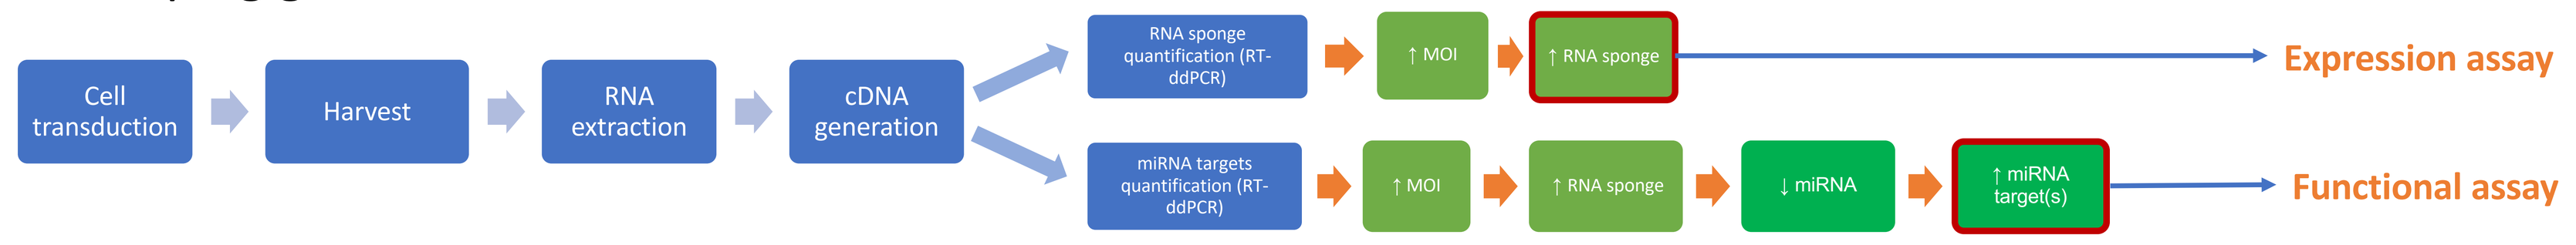

Cell transduction
Harvest
RNA extraction
cDNA generation
RNA sponge quantification (RT-ddPCR)
↑ MOI
↑ RNA sponge
Expression assay
miRNA targets quantification (RT-ddPCR)
↑ MOI
↑ RNA sponge
↓ miRNA
↑ miRNA target(s)
Functional assay

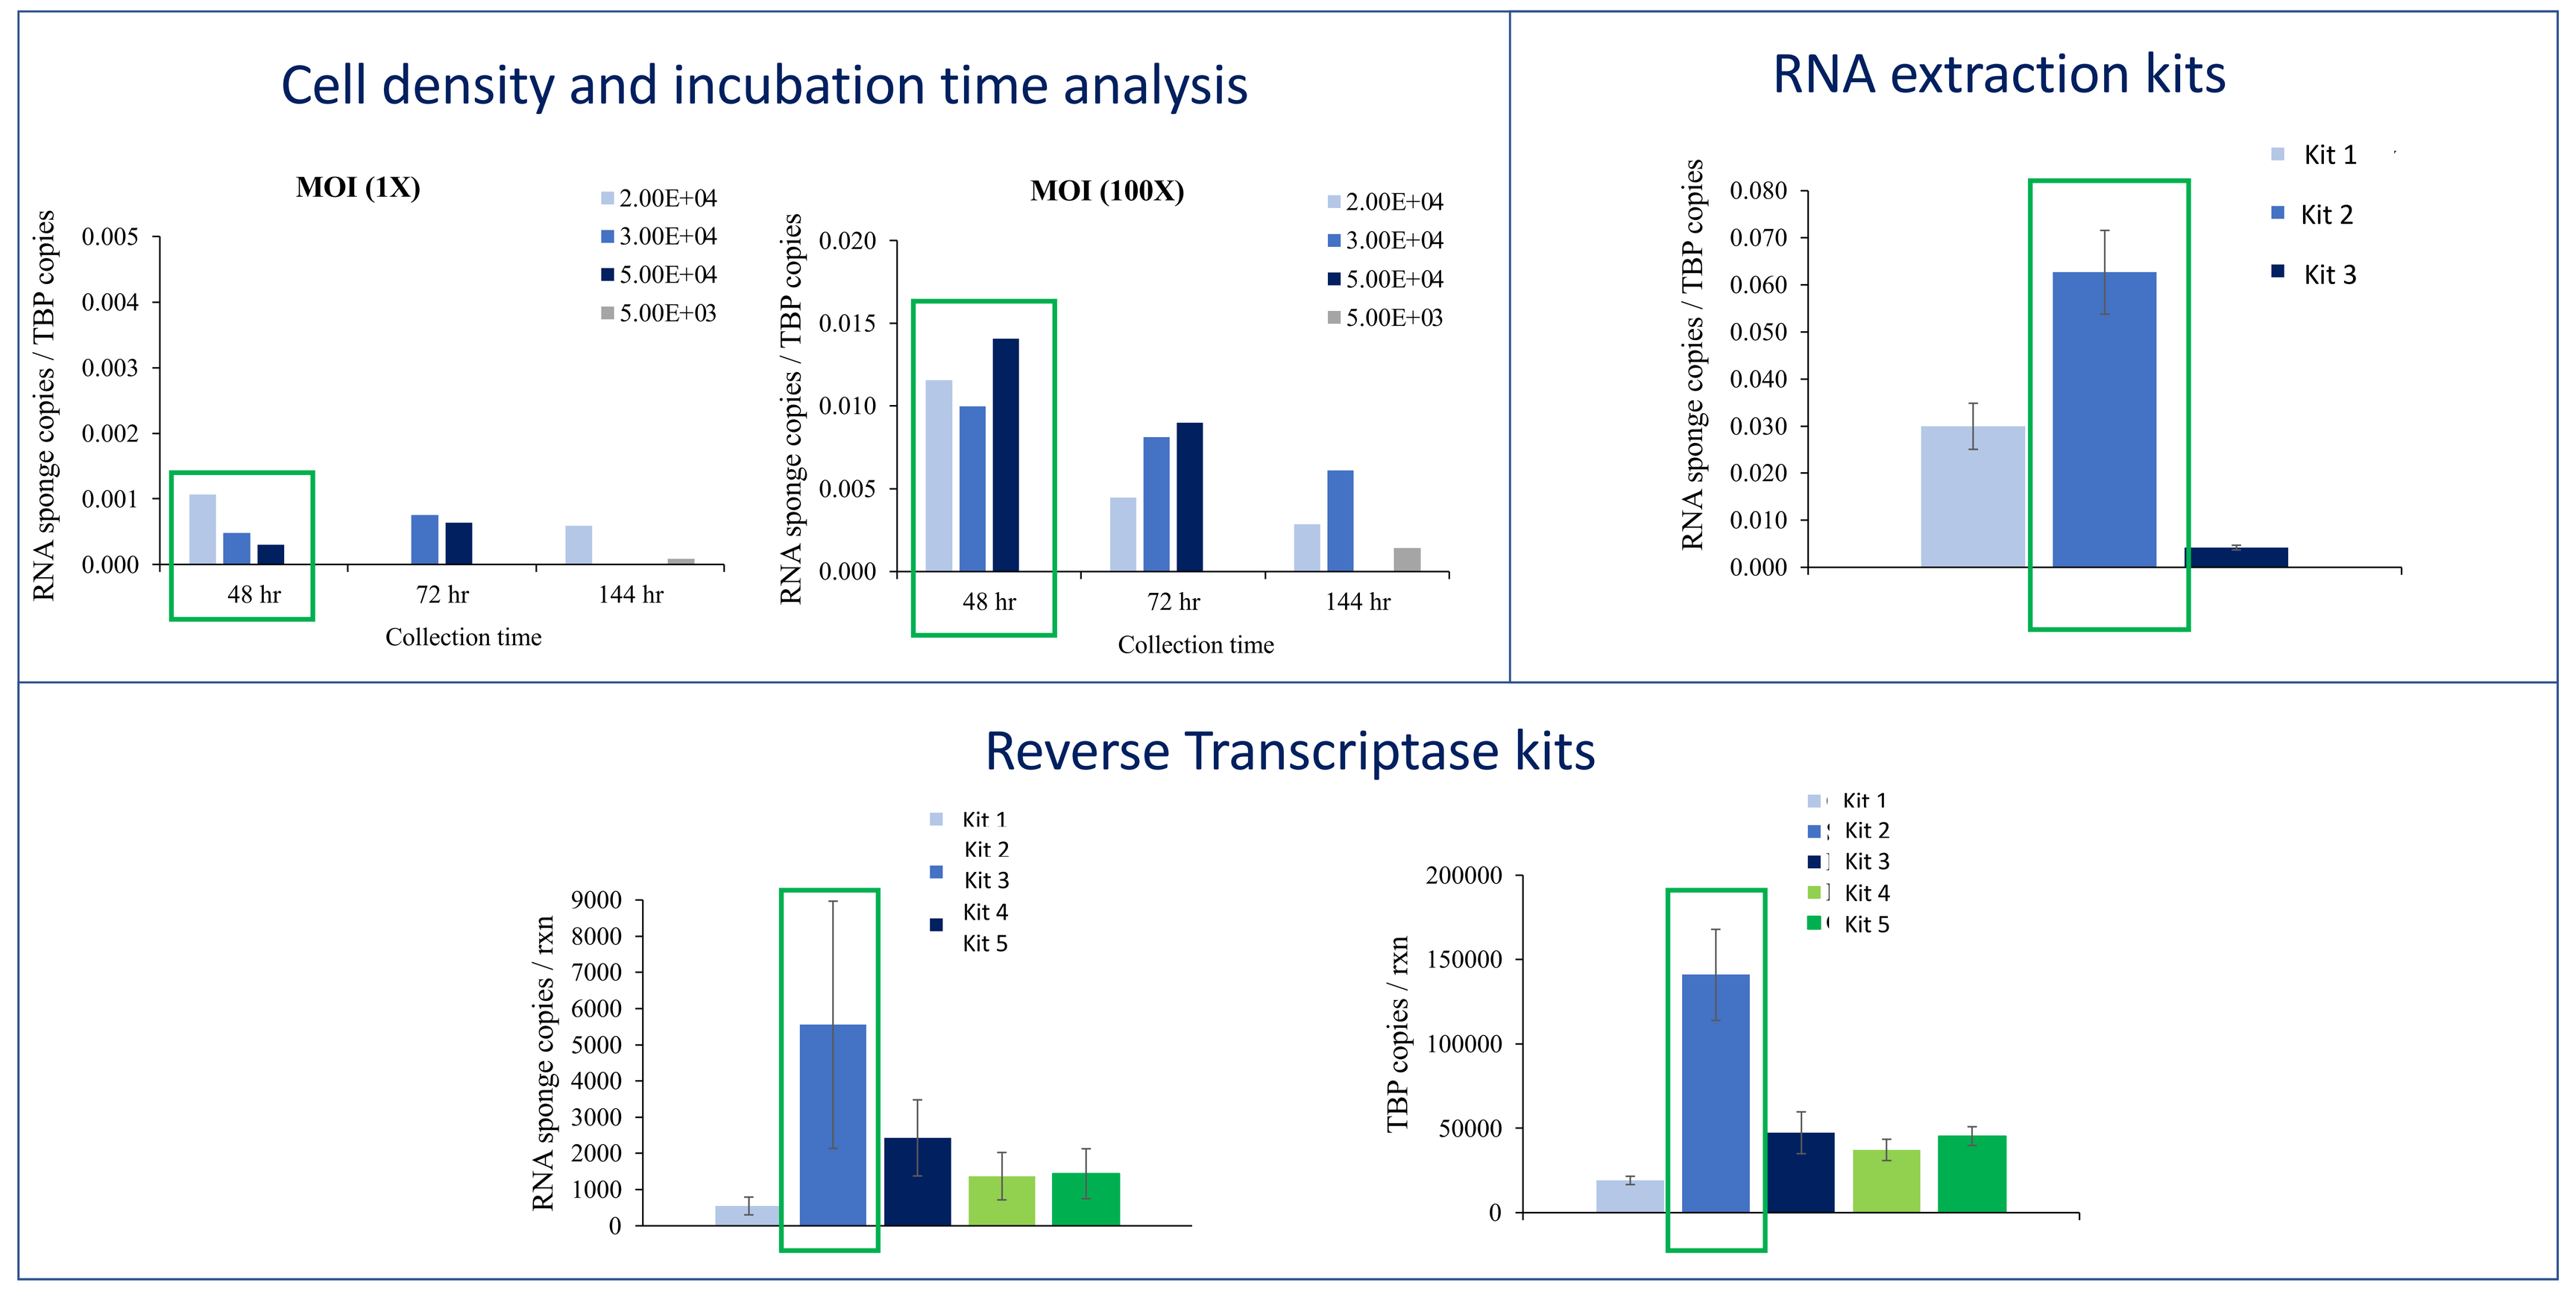

Cell density and incubation time analysis
MOI (1X)
MOI (100X)
2.00E+04
3.00E+04
5.00E+04
5.00E+03
RNA sponge copies / TBP copies
48 hr
72 hr
144 hr
Collection time
RNA extraction kits
Kit 1
Kit 2
Kit 3
Reverse Transcriptase kits
Kit 1
Kit 2
Kit 3
Kit 4
Kit 5
RNA sponge copies / rxn
TBP copies / rxn

FIGURE 2: Cells were transduced with the vector at two different MOIs for cell density and incubation time analysis. For RNA extraction kits and RT kits tests, cells were transfected at the higher MOI only. The conditions selected are highlighted in green rectangles.

Process Development

Suspension vs. Adherent

First in human trials require getting to clinic quickly and cost effectively and often utilize bench processes that are adherent and non-scalable. Development of late-stage processes therefore require comparability studies that can be time consuming and regulatorily challenging. To overcome these challenges and allow for flexibility between early and late-stage processes, a suspension cell line was tested in both adherent (T-flask) and suspension (shake flask) small-scale conditions. The data confirm that these cells performed well under both conditions and within industry expectations.

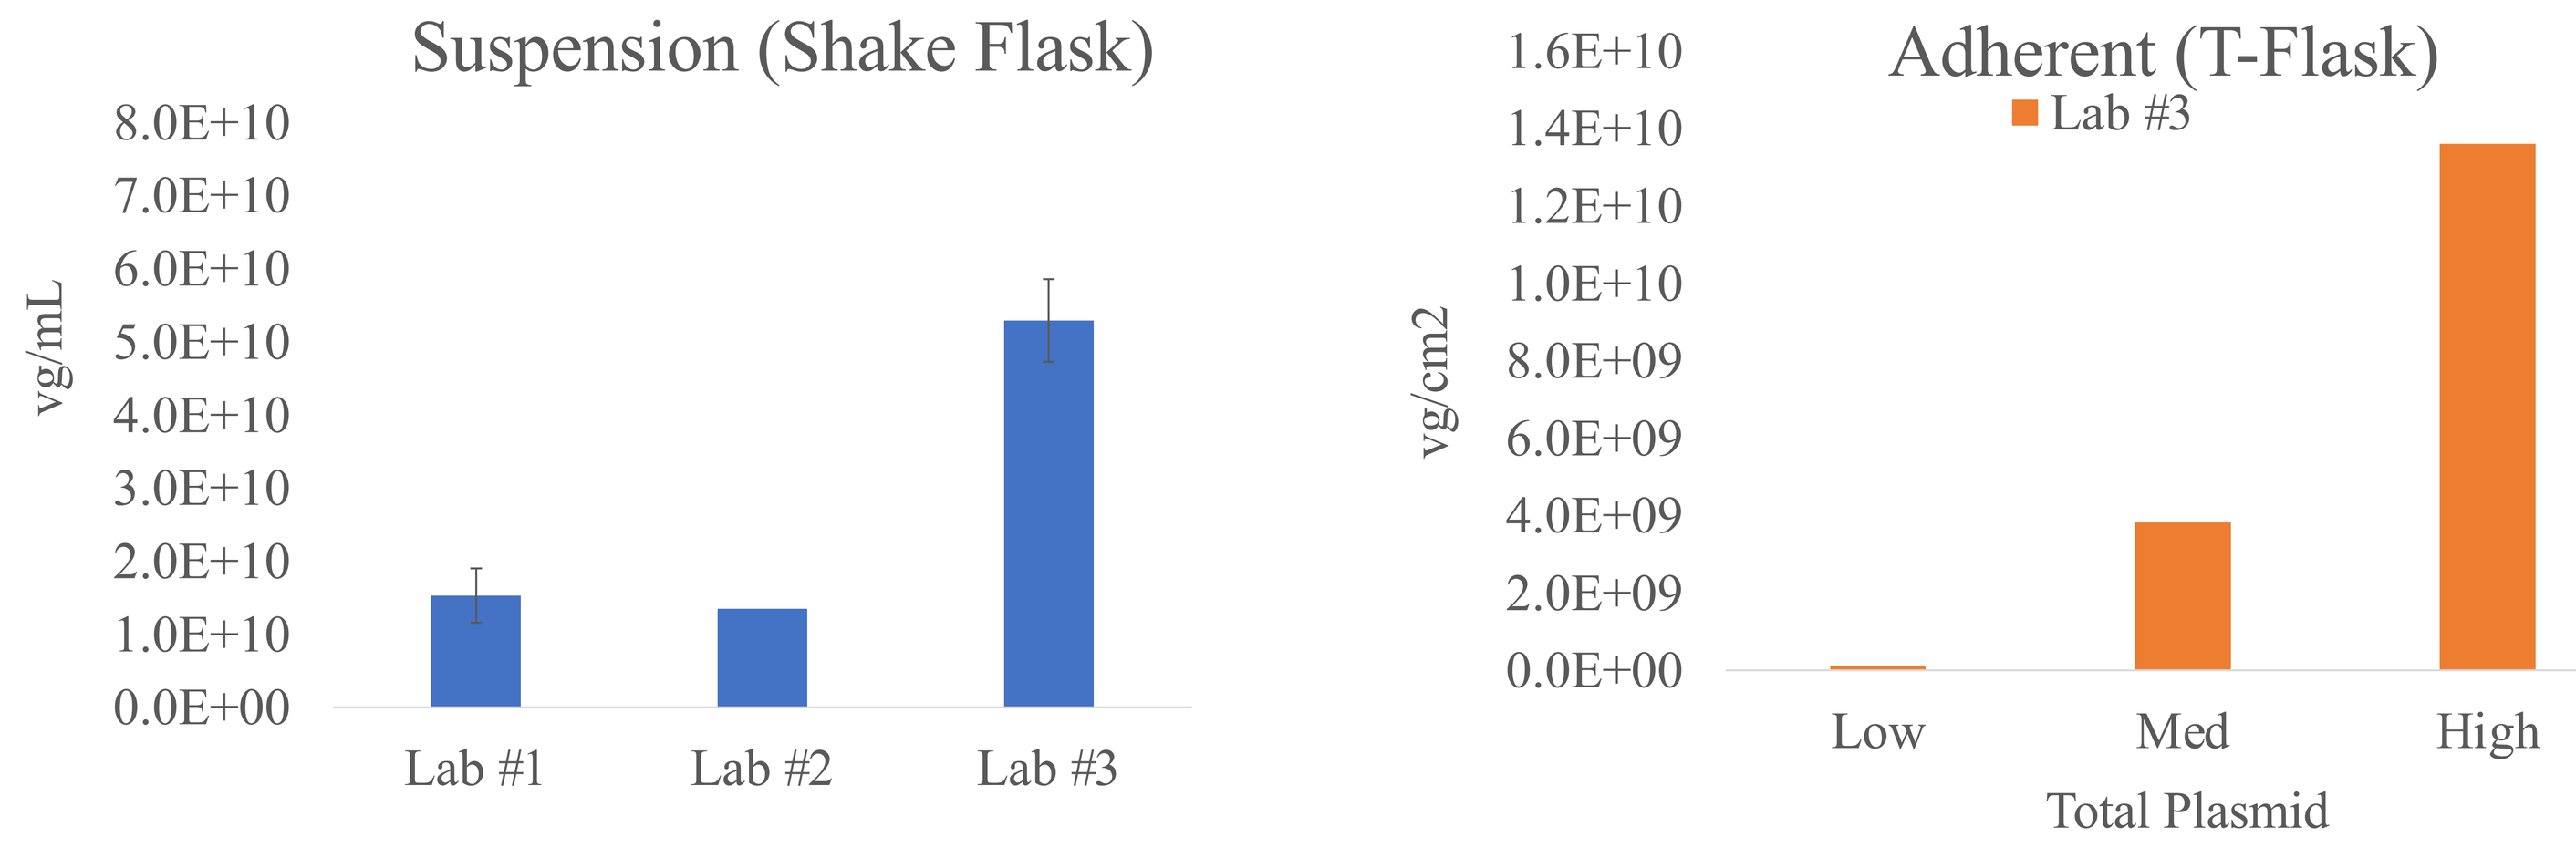

Suspension (Shake Flask)
vg/mL
Lab #1
Lab #2
Lab #3
Adherent (T-Flask)
Lab #3
vg/cm2
Low
Med
High
Total Plasmid

FIGURE 3: Suspension HEK293 cells were evaluated in shake flasks and T-flasks under platform conditions (1:1 PEI:DNA, 1:1:1 mass ratio of Help:RepCap:Transgene). Harvest was performed four days post transfection by lysing cells with Tween-20. For suspension, data were collected from three independent labs to yield comparable data (higher titers obtained from Lab #3 may be attributed to variability in assay conditions although the same primer/probe sets were used). Suspension cells can be transfected under adherent conditions and show appreciable dose response curves (increased productivity with increase in total plasmid DNA). Additional optimization is ongoing to scale these conditions.

Scale-up

The suspension process was transferred into a 50L stir tank bioreactor (ThermoFisher Hyperforma) using the baseline, unoptimized conditions used in shake flasks. Data indicate that this process is scalable and produces comparable titers.

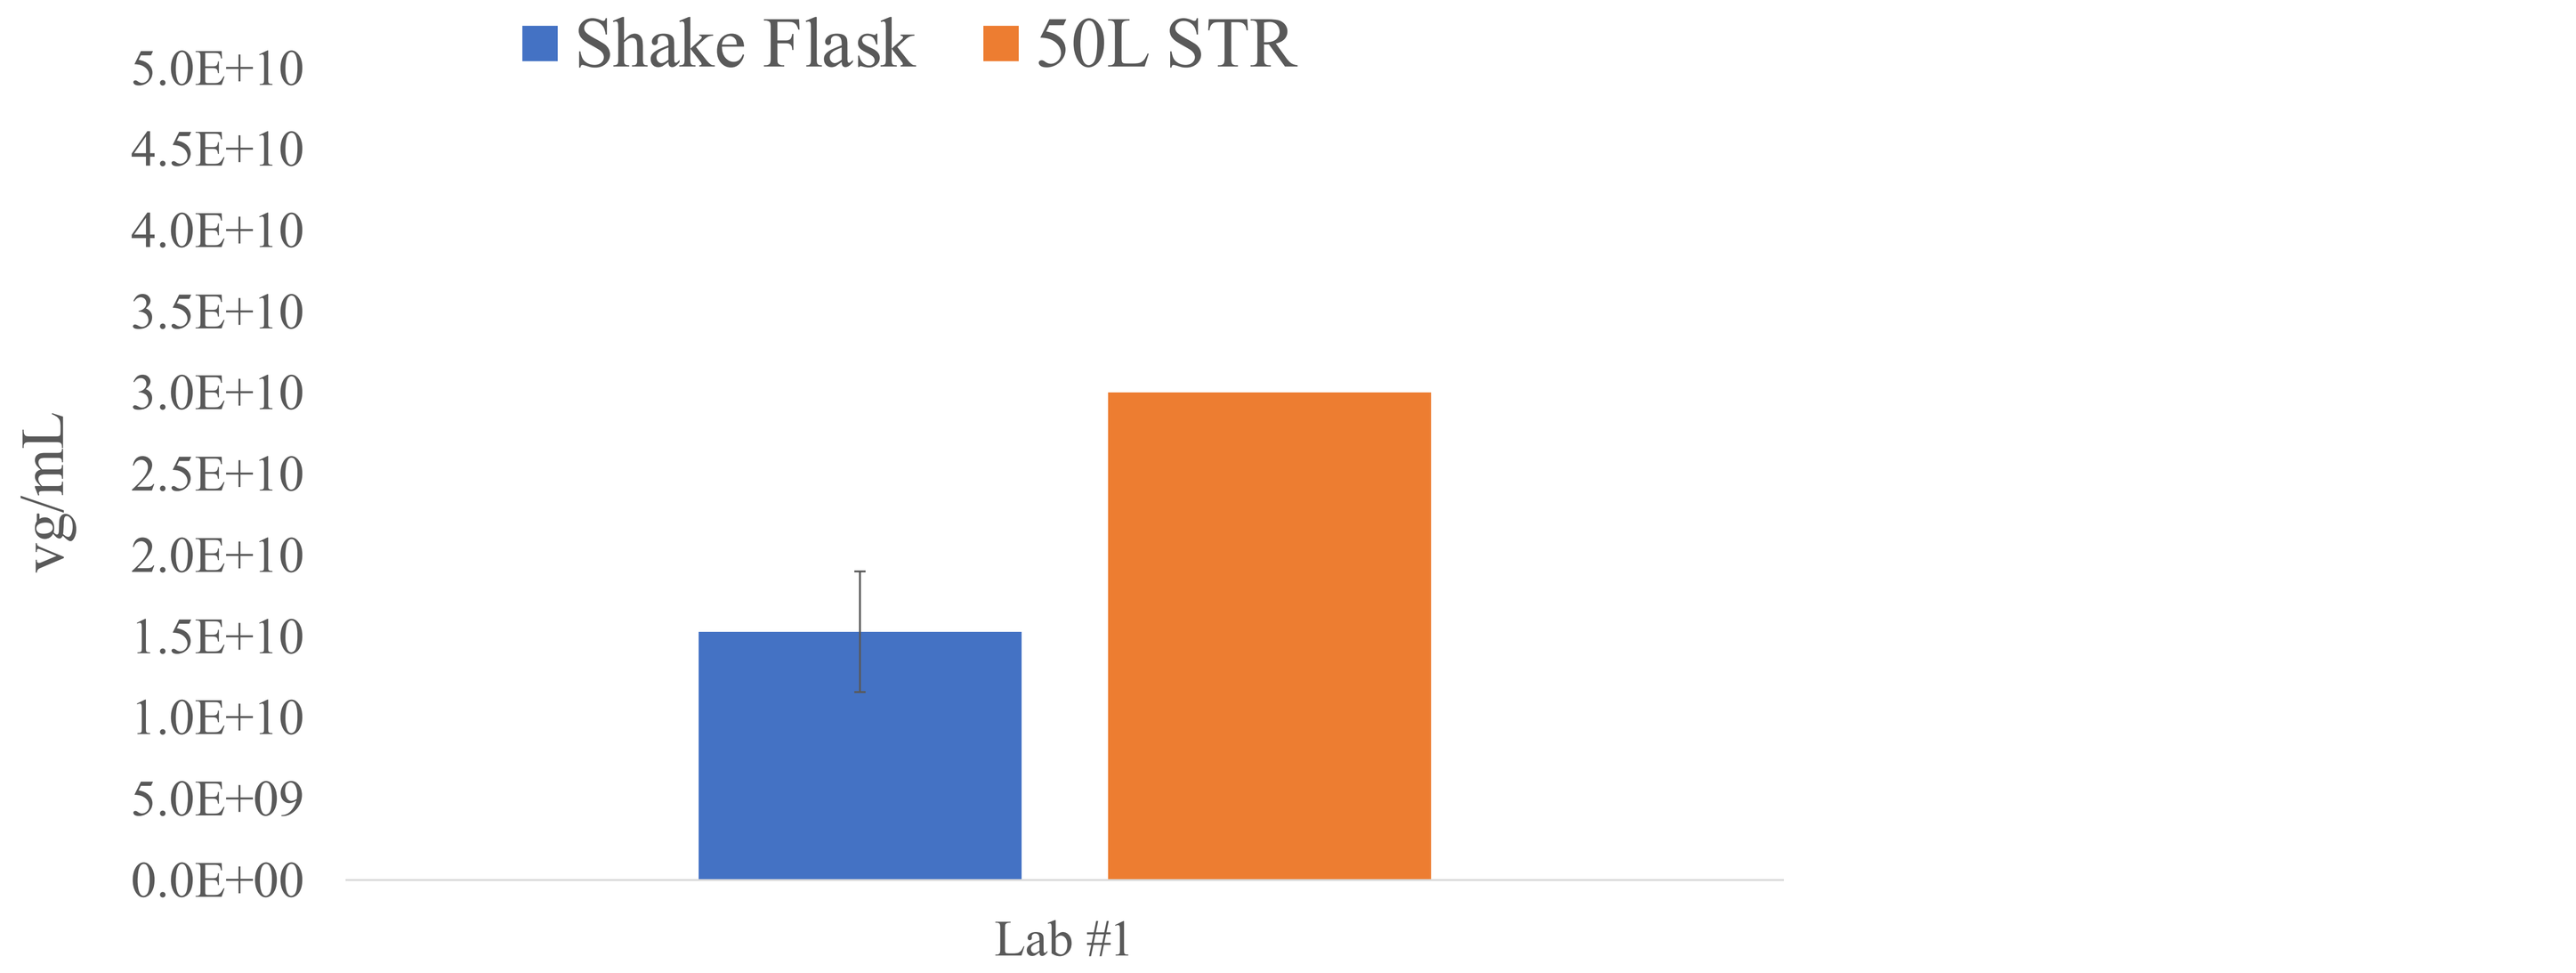

Shake Flask
50L STR
vg/mL
Lab #1

FIGURE 4: The suspension process was successfully scaled to a 50L bioreactor with comparable productivity. While these titers meet industry expectations for AAV9, further optimization of parameters are being performed to improve productivity.

Cell Lines

Two suspension cell lines were tested using unoptimized conditions in shake flasks. The same transgene and rep/cap plasmids were used with both cell lines, but a different helper plasmid was used with cell line B. The data are comparable between labs, cell lines and helper plasmids given the unoptimized conditions.

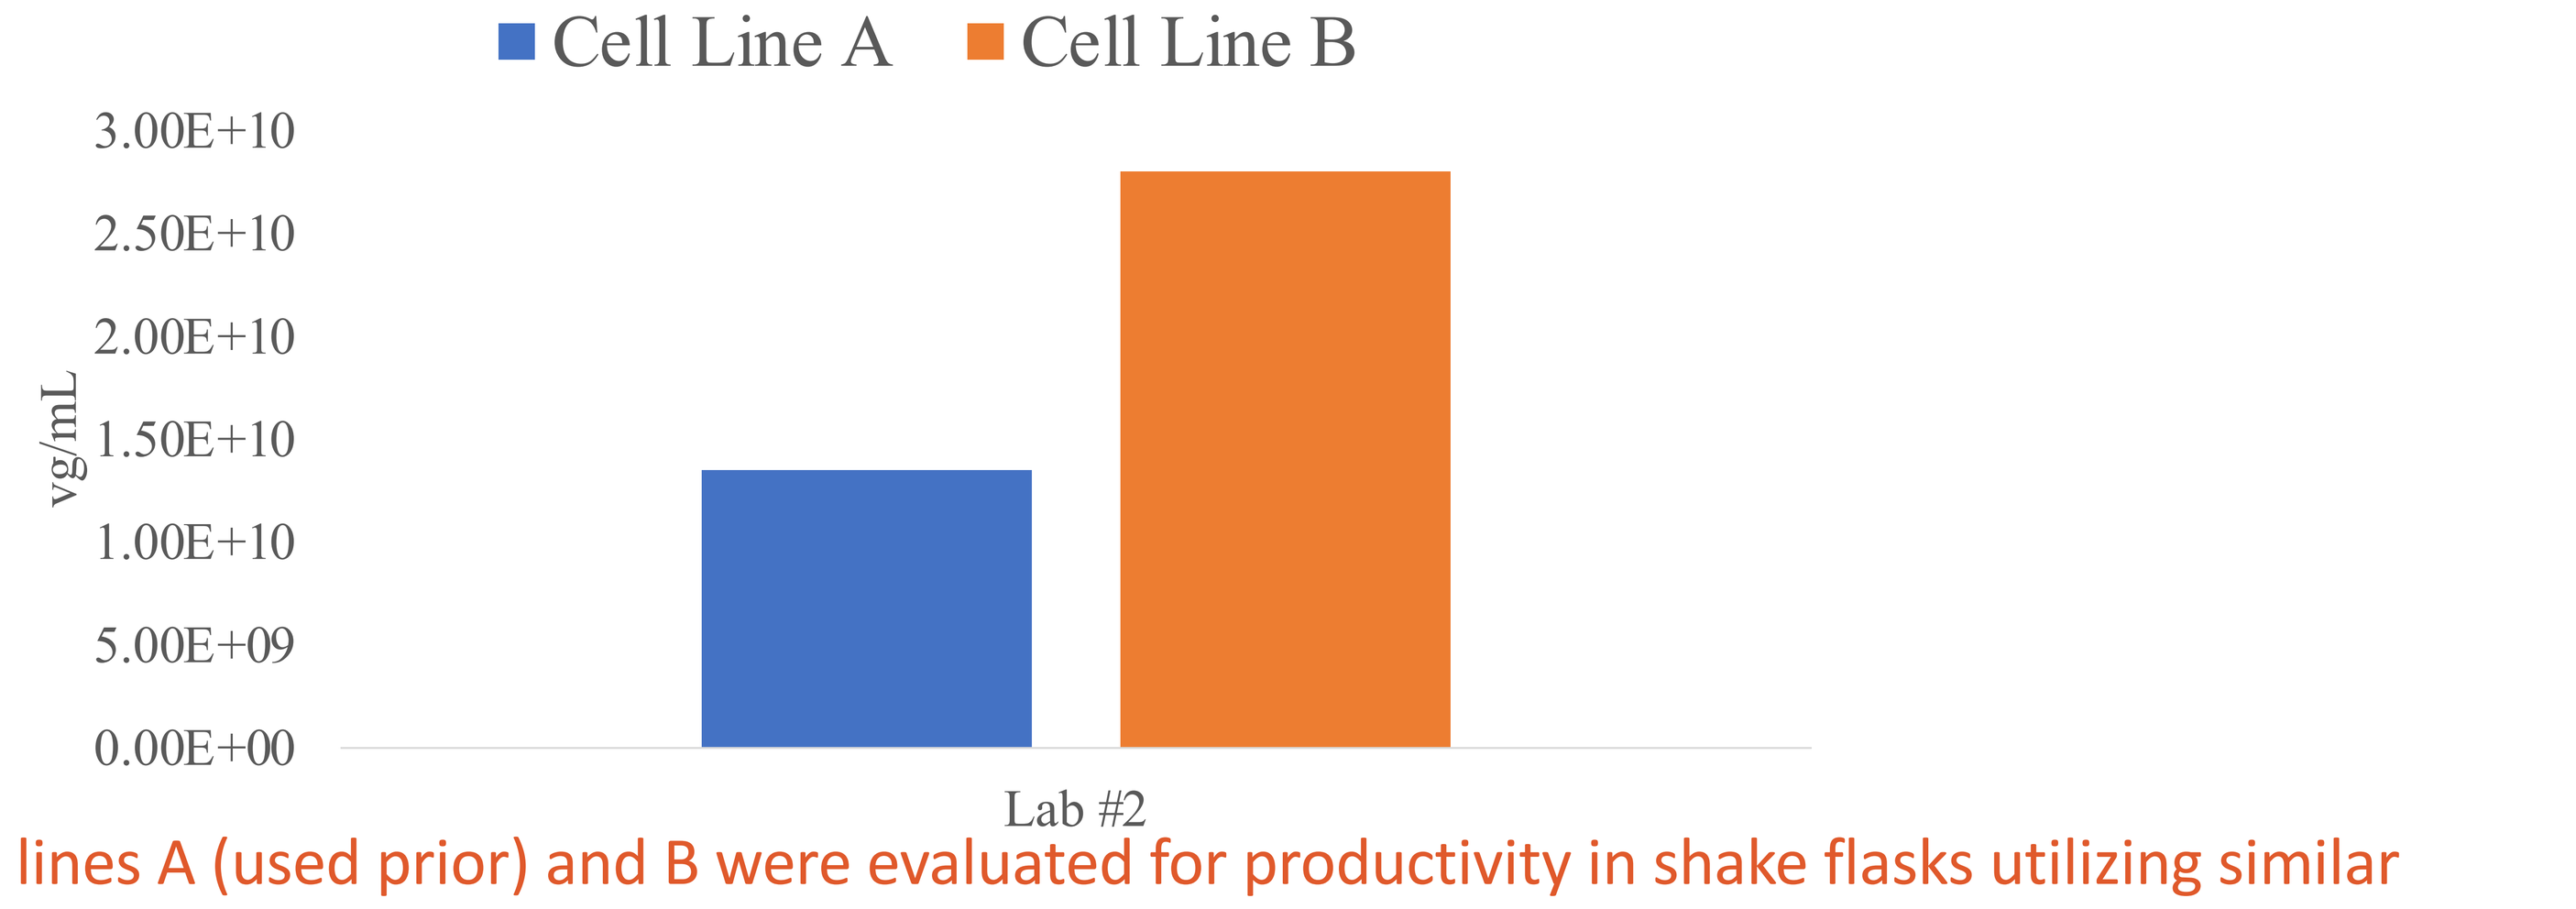

Cell Line A
Cell Line B
vg/mL
Lab #2

FIGURE 5: Cell lines A (used prior) and B were evaluated for productivity in shake flasks utilizing similar parameters for transfection (except for the helper plasmid) and harvest. Comparable data were obtained, but cell line B trended higher in titer.

Conclusions

- Development and optimization for dosing (vg) and potency assays are critical to ensure optimal process development and continued robustness of process and critical quality attributes (CQA)
- Development and optimization of an upstream process are critical and must meet criteria for productivity and scalability with a line of sight to regulatory and economic challenges for commercialization
- We have developed an upstream process that is flexible, versatile and scalable, but still undergoing continuous process improvements for productivity.
- Downstream process development and optimization is currently ongoing.

Acknowledgments

This work was performed in partnership with several CDMOs and CROs. A portion of the laboratory work for this study was performed in conjunction with The Meyer Laboratory at Nationwide Children's Hospital. We thank our colleagues at Alcyone Therapeutics and Nationwide Children's Hospital for their support with this poster.